Center for Alternatives to Animal in Testing

Higher-throughput, allometrically-scaled, two-chamber liver-organ co-culture system for toxicity testing.

Blanche C. Ip[1] (blanche_ip@brown.edu), Samantha J. Madnick[1], Sophia Zheng[1], Hui Li[1], Susan J. Hall[1], Suzanne Martin[2], Richard Cubberley[2], Jeffrey R. Morgan[1], and Kim Boekelheide[1]

[1]Department of Pathology and Laboratory Medicine at Brown University, RI, United States; [2]Safety and Environmental Assurance Centre, Unilever, United Kingdom

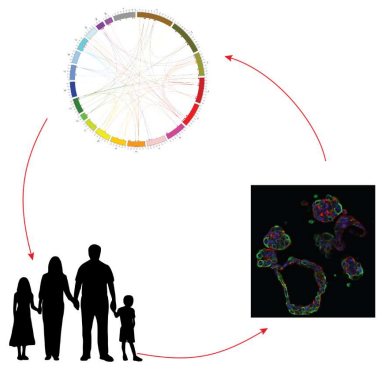

Objectives:

Ingested chemicals and drugs often undergo hepatic xenobiotic metabolism and generate a spectrum of metabolites. These metabolites can exhibit greater or lesser toxicity than the parent compounds for a given biological target through hepatic bioactivation and bioinactivation, respectively. Our **research goals are to:**

1. Design and fabricate two-chamber liver-organ co-culture system in a higher-throughput 96-well format for the determination of toxicity on target tissues in the presence of physiologically relevant human liver metabolism. This hydrogel system within a single well consists an outer ring-shaped trough to culture human liver cells as three-dimension (3D) microtissues, and a central well to house target cells in 2D or 3D. The system is allometrically scalable to produce sufficient quantities of metabolites to simulate the in vivo exposure setting.
2. Characterize 3D liver microtissues formation kinetics and physiology made with HepaRG®, then optimize hepatic metabolism and function.
3. Demonstrate the inert nature of the hydrogel component of the two-chamber system, where chemicals introduced and generated from hepatic metabolism can rapidly diffuse between the two chambers, and that chemicals do not adsorb onto the hydrogel.

Methods:

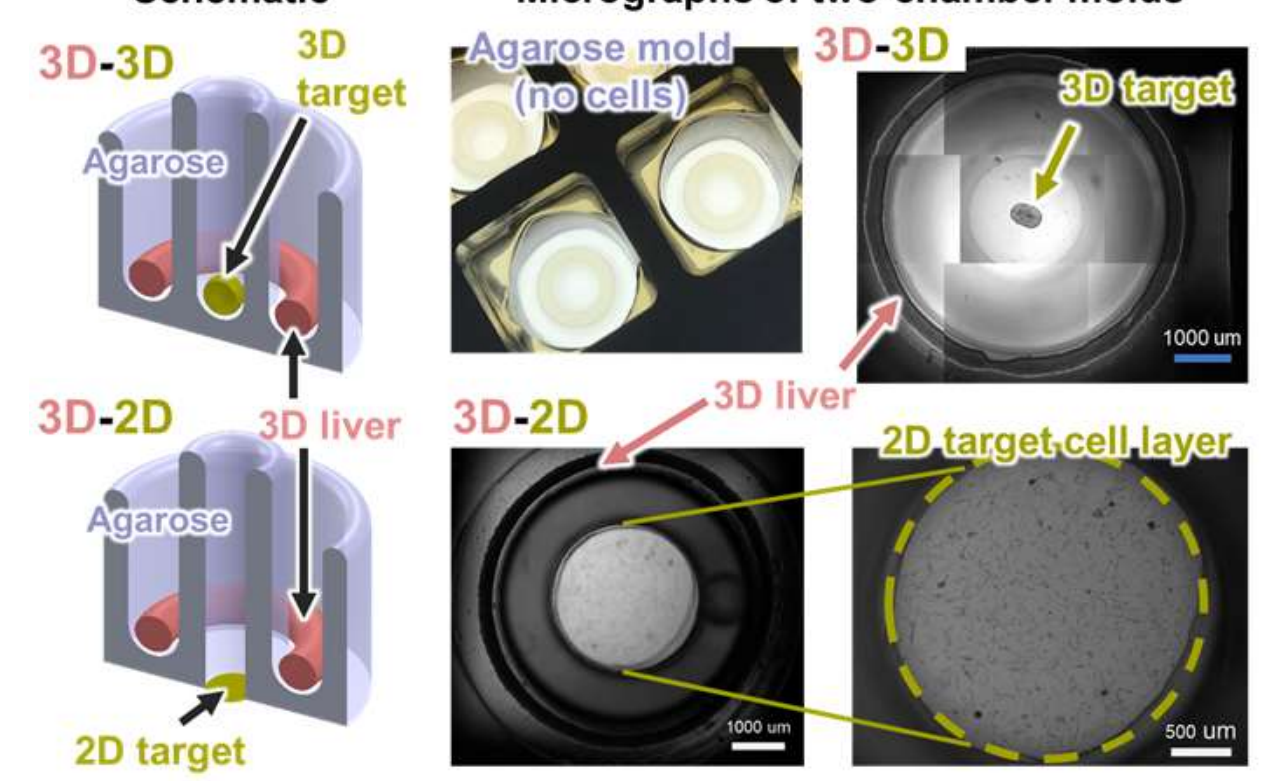
Schematic
Micrographs of two-chamber molds
3D-3D
3D target
Agarose
Agarose mold (no cells)
3D-3D
3D target
1000 um
3D-2D
3D liver
3D-2D
3D liver
2D target cell layer
Agarose
2D target
1000 um
500 um

Fig 1: Two-chamber agarose system. Two versions of the system have been developed consisting of a 3D liver microtissue(s) with either a 3D target microtissue (3D-3D) or a 2D monolayer of target cells (3D-2D).

HepaRG™ culturing as 3D microtissues:

HepaRG cells (Lonza) were thawed and seeded into 2% agarose molds and maintained in Williams E medium with HepaRG supplements (MHTAP, MHPIT or MHMET; Lonza), GlutaMax (ThermoFisher) and 1% Pen/Strep. Media was changed every 1 or 2 days for 10 or 17 days. Selected tissues were fixed in 10% formalin, paraffin embedded and sectioned for histological analysis of general morphology and hepatic biomarkers, or snapped frozen to evaluate hepatic Phase I cytochrome P450 (*Cyp1a2, Cyp2b6, Cyp3a4*) gene expression. Selected tissues were treated with different chemicals to examine hepatic Phase I metabolism of HepaRG 3D microtissues. N = 3 to 10 per group.

Results:

➢ **HepaRG forms stable 3D microtissues in two-chamber system: Live cell imaging and histology.**

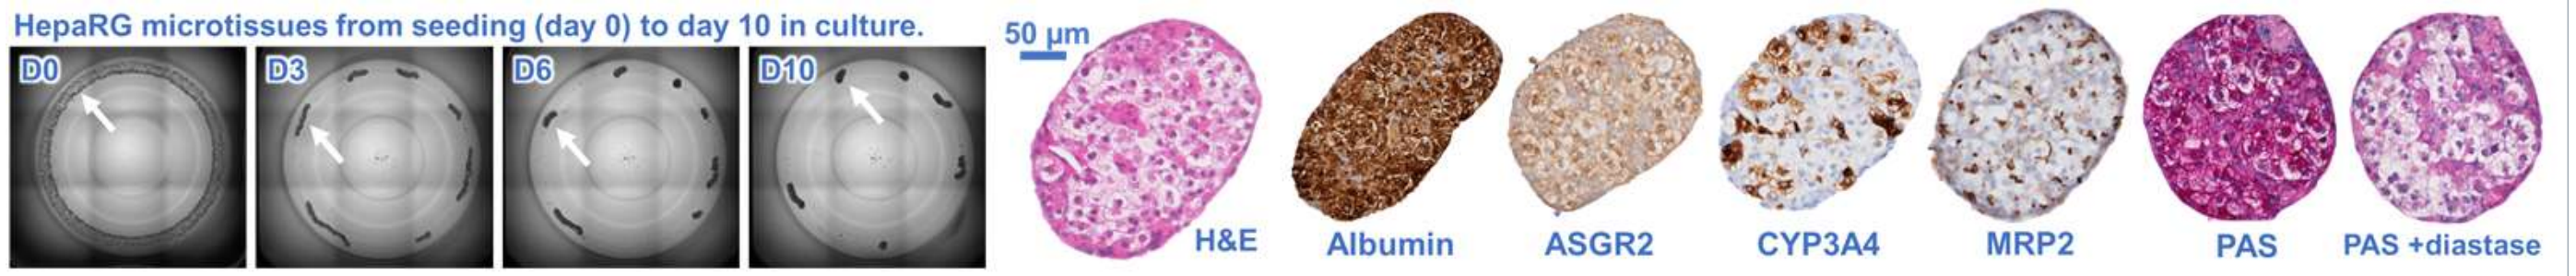
HepaRG microtissues from seeding (day 0) to day 10 in culture.
D0
D3
D6
D10
50 µm
H&E
Albumin
ASGR2
CYP3A4
MRP2
PAS
PAS +diastase

Fig 2: Live cell brightfield imaging with a PerkinElmer Opera Phenix® high-content screening system showed the HepaRG cells (50K) in the two-chamber system formed stable 3D microtissues (arrows) that compacted over the course of 10 days. 10-day old HepaRG microtissues were formalin-fixed, paraffin-embedded and sectioned for histological analyses. Hematoxylin and eosin (H&E) staining shows HepaRG cells in 3D microtissues have abundant cytoplasm. Immunohistochemical staining (in brown) reveals tissue expression of albumin, asialoglycoprotein receptor (ASGR2) for glycoprotein uptake, Phase I cytochrome P450 (CYP3A4) enzyme, and Multidrug resistance-associated protein 2 (MRP2) transporter for biliary excretion. Periodic acid–Schiff (PAS) stain with or without diastase enzyme that breaks down glycogen supports that HepaRG microtissues synthesize and store glycogen.

➢ **HepaRG microtissues have differential hepatic Phase I gene expressions and protein functions that are dependent on maturation time and media supplement.**

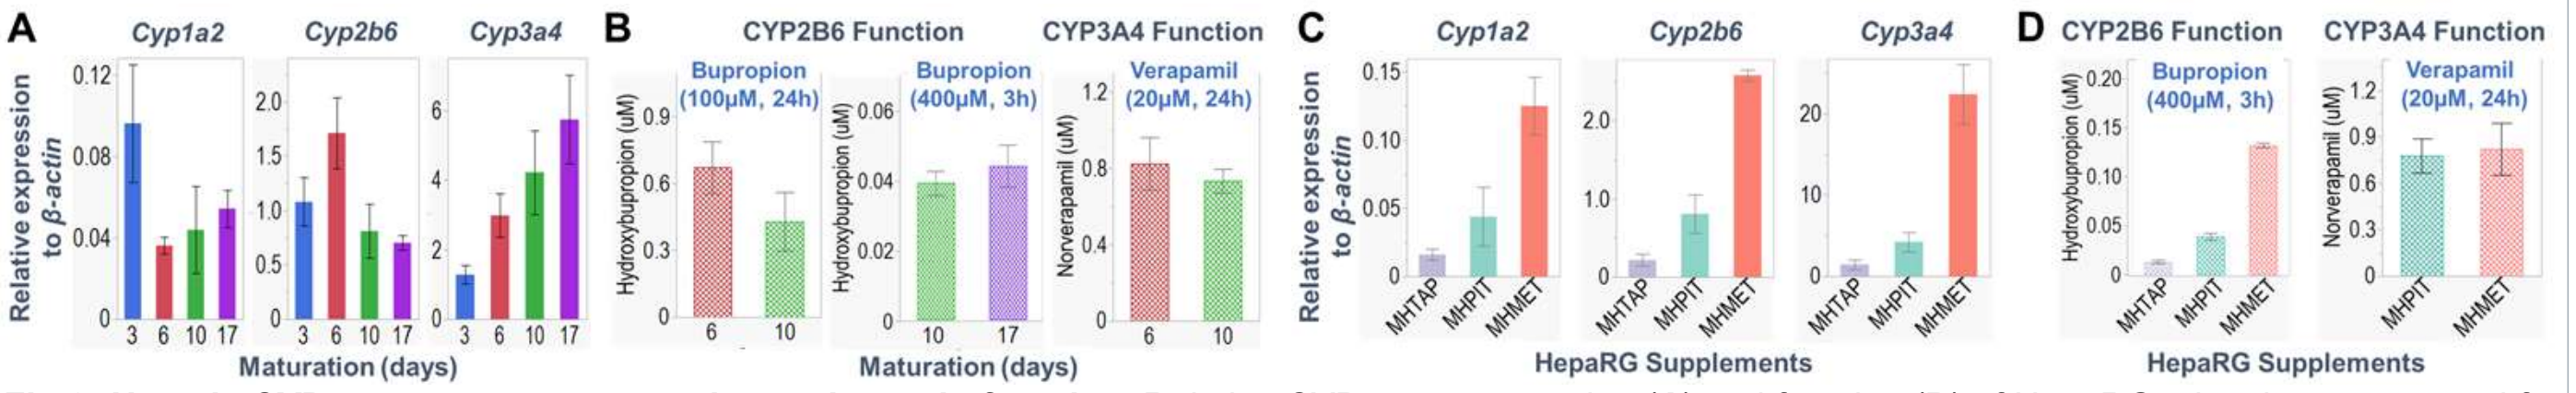
A
Cyp1a2
Cyp2b6
Cyp3a4
Relative expression to β-actin
Maturation (days)
B
CYP2B6 Function
CYP3A4 Function
Bupropion (100µM, 24h)
Bupropion (400µM, 3h)
Verapamil (20µM, 24h)
Hydroxybupropion (uM)
Norverapamil (uM)
Maturation (days)
C
Cyp1a2
Cyp2b6
Cyp3a4
Relative expression to β-actin
MHTAP
MHPIT
MHMET
HepaRG Supplements
D
CYP2B6 Function
CYP3A4 Function
Bupropion (400µM, 3h)
Verapamil (20µM, 24h)
Hydroxybupropion (uM)
Norverapamil (uM)
HepaRG Supplements

Fig 3: Hepatic CYP enzyme gene expression and protein function. Relative CYP gene expression (**A**) and function (**B**) of HepaRG microtissues matured for 3, 6, 10 or 17 days in MHPIT after seeding. Relative CYP gene expression (**C**) and function (**D**) of HepaRG microtissues matured for 10 days after seeding in MHTAP, MHPIT or MHMET (contains 0%, 0.5% or 1.67% DMSO, respectively). Data = mean ± SD.

➢ **Agarose does not adsorb compounds tested: phenacetin, albumin and estradiol.**

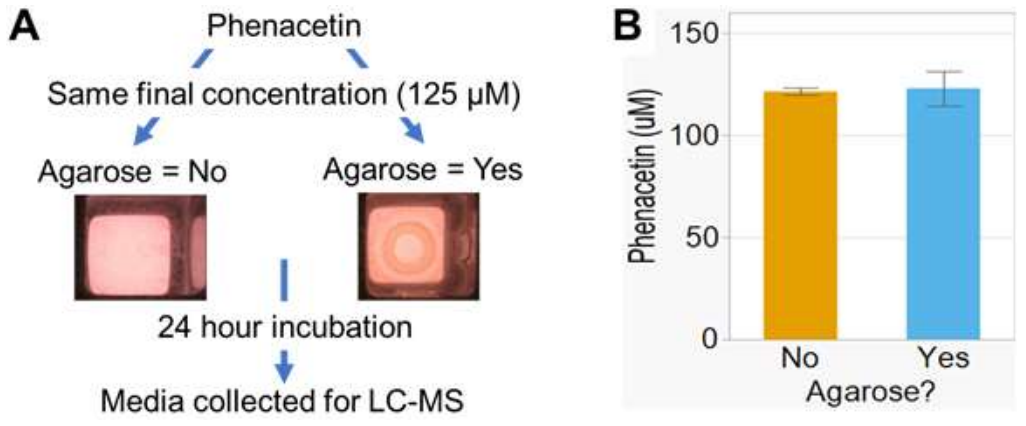
A
Phenacetin
Same final concentration (125 µM)
Agarose = No
Agarose = Yes
24 hour incubation
Media collected for LC-MS
B
Phenacetin (uM)
No
Yes
Agarose?

Fig 4: Agarose does not absorb phenacetin. Methods for examining phenacetin adsorption onto agarose (**A**). Phenacetin concentration by LC-MS after 24h incubation in well with or without the two-chamber agarose system (**B**). Data = mean ± SD.

➢ **Diffusion of chemicals occurs rapidly within the two-chamber system.**

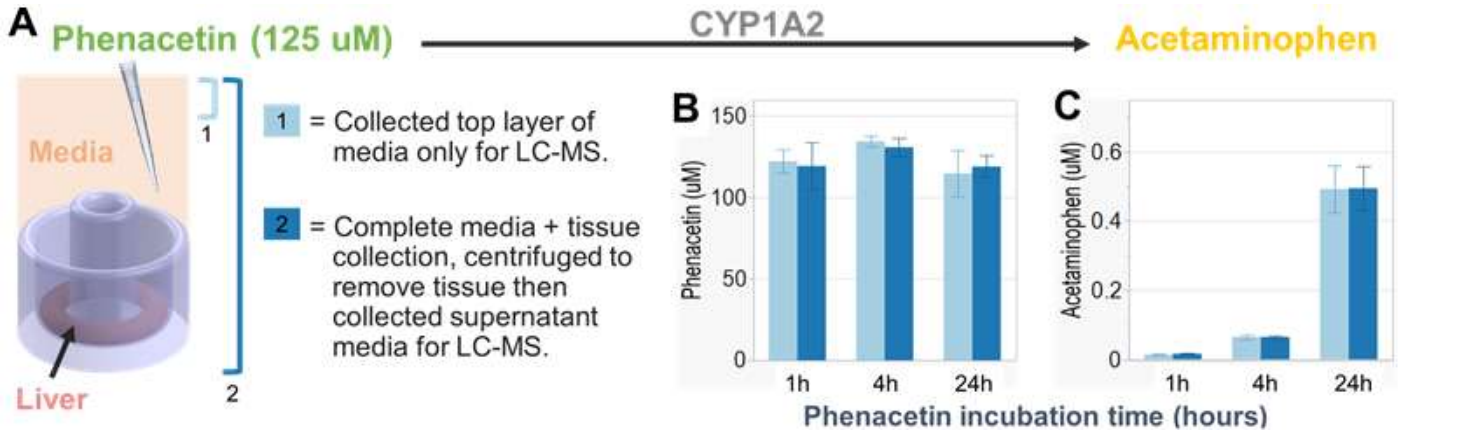
A
Phenacetin (125 uM)
CYP1A2
Acetaminophen
Media
Liver
1 = Collected top layer of media only for LC-MS.
2 = Complete media + tissue collection, centrifuged to remove tissue then collected supernatant media for LC-MS.
B
Phenacetin (uM)
C
Acetaminophen (uM)
1h
4h
24h
Phenacetin incubation time (hours)

Fig 5: Compounds diffusion occurs rapidly. To examine phenacetin and acetaminophen diffusion in two-chamber agarose system, HepaRG microtissues were incubated in phenacetin for 1, 4 or 24h, followed by media collection with two different media removal methods for LC-MS analysis (**A**). Phenacetin (**B**) and acetaminophen (**C**) concentrations were comparable between media removal methods. Data = mean ± SD.

Conclusion and Future Directions:

Conclusion: Our liver-organ co-culture platform provide critical and higher-throughput testing for metabolism-dependent bioactivity of chemicals, to better recapitulate and predict potential human health hazards of compounds.

Future directions: The next steps are case studies of: 1) pro-estrogenic activation by HepaRG microtissues of chemicals that alter proliferation and viability of breast cancer cells; 2) metabolic modulation of CALUX® reporters.

Acknowledgements:

Funding: NIH-NIEHS (U01 ES028184), Unilever, UK and the generous gift by Donna McGraw Weiss ('89) and Jason Weiss. We thank Corteva, Inc. members (Kamin Johnson, Wei Chen, Andrew Bowling, Heather Pence and Lori L Breitweiser) for their support. We also thank Dr. Steven Ferguson (NIEHS) for his advice on the culturing of HepaRG cells in 3D.